Iota-Carrageenan is a potent inhibitor of rhinovirus infection

A.Grassauer[1,2], R. Weinmuellner[1], C. Meier[1,2], E. Prieschl-Grassauer[1,2] and H. Unger[2]

[1]Marinomed Biotechnologie GmbH, Veterinaerplatz 1/HA, A-1210 Vienna, Austria
[2]Laboratory of Tropical Veterinary Medicine, Veterinary University Vienna, Veterinaerplatz 1, A-1210 Vienna, Austria

Background: Human rhinoviruses (HRVs) are the predominant cause of common cold. In addition, HRVs are implicated in the worsening of COPD and asthma, as well as the loss of lung transplants. Despite significant efforts, no anti-viral agent is approved for the prevention or treatment of HRV-infection.

Results: In this study we demonstrate that Iota-Carrageenan, a sulphated polysaccharide derived from red seaweed, is a potent anti-rhinoviral substance in-vitro. Iota-Carrageenan reduces HRV growth and inhibits the virus induced cythopathic effect of infected HeLa cells. In addition, Iota-Carrageenan effectively prevents the replication of HRV1A, HRV2, HRV8, HRV14, HRV16, HRV83 and HRV84 in primary human nasal epithelial cells in culture. The data suggest that Iota-Carrageenan acts primarily by preventing the binding or the entry of virions into the cells.

Conclusion: Since HRV infections predominately occur in the nasal cavity and the upper respiratory tract, a targeted treatment with a product containing Iota-Carrageenan is conceivable. Clinical trials are needed to determine whether Iota-Carrageenan-based products are effective in the treatment or prophylaxis of HRV infections.

Carrageenan promotes cell survival after HRV2 infection and reduces viral particle production

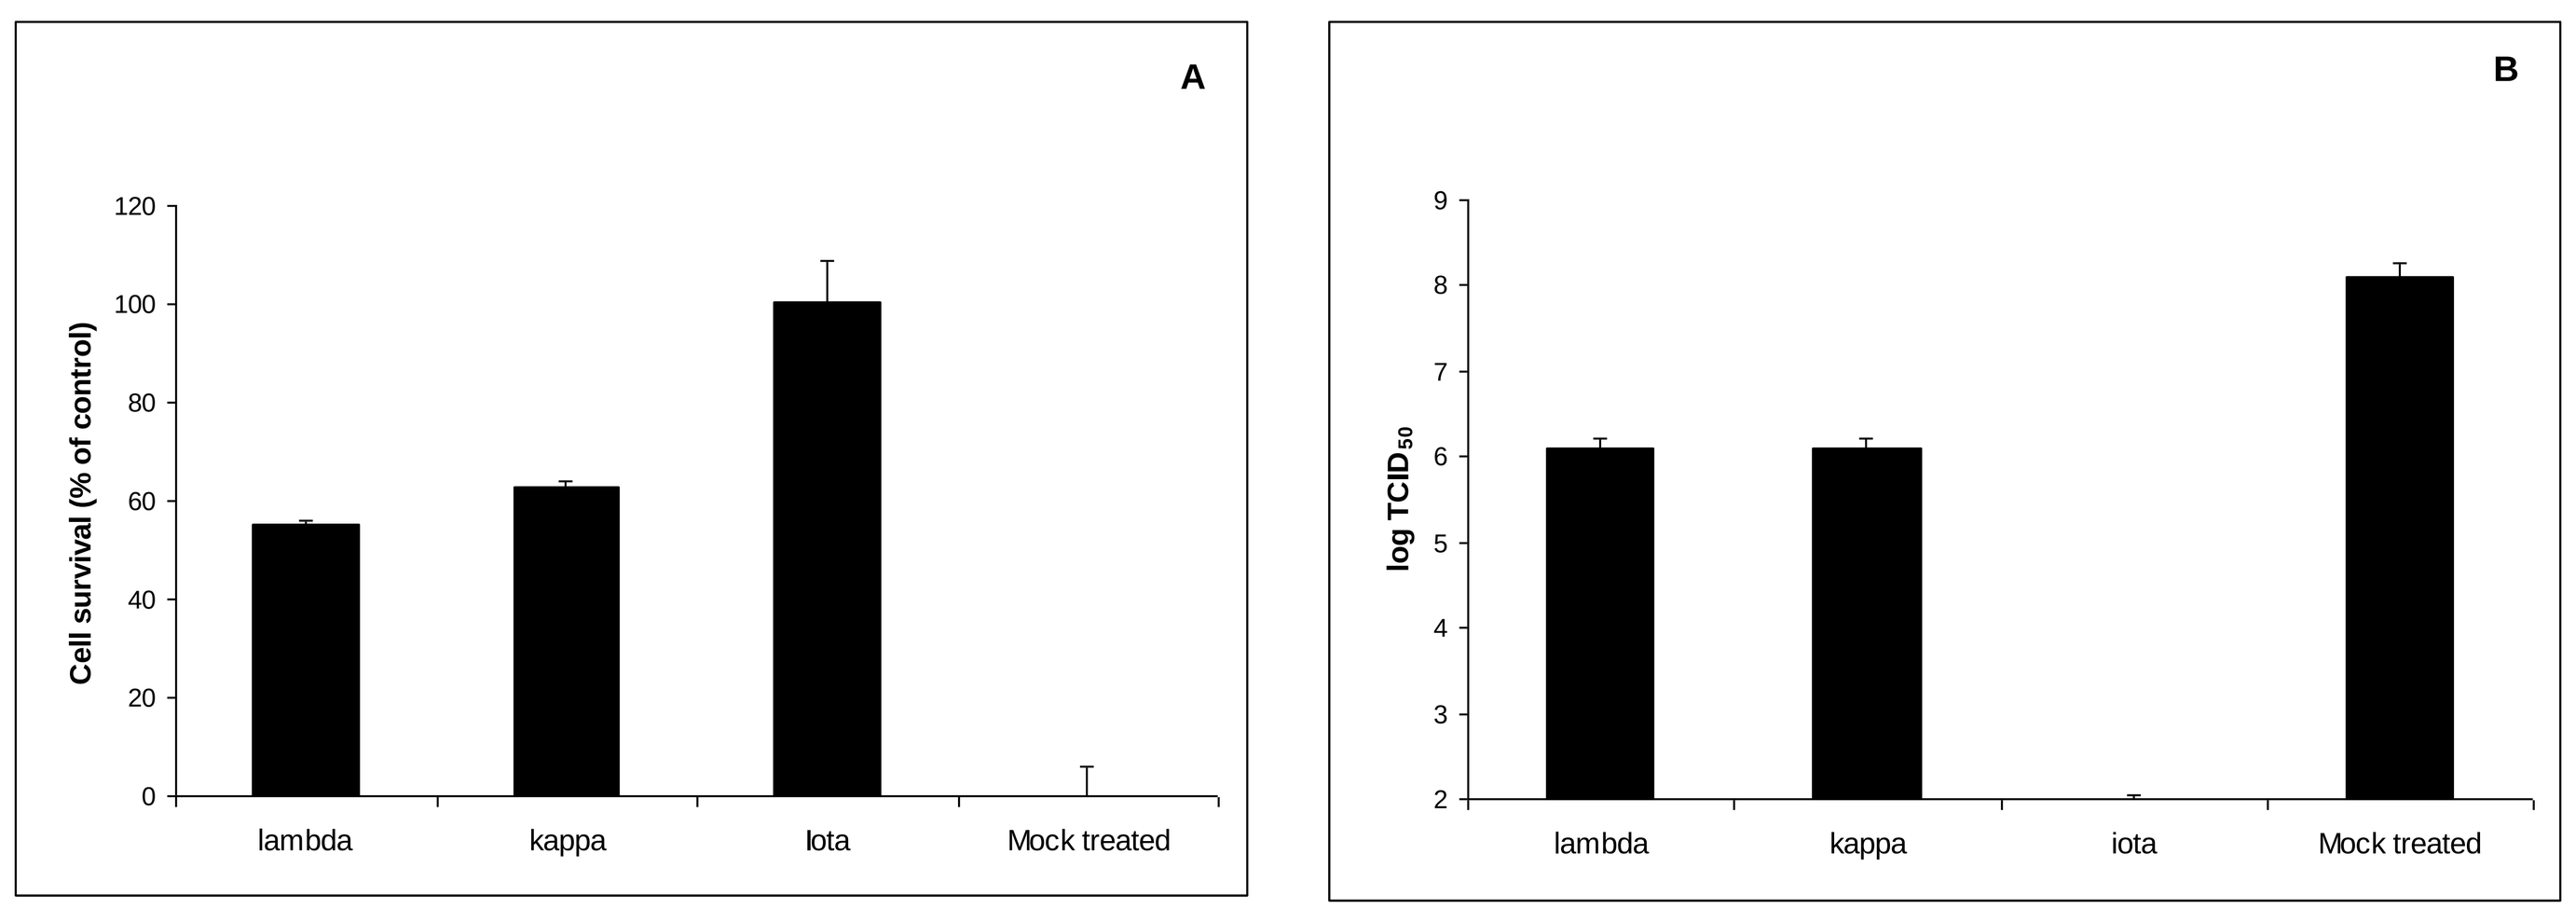
A
Cell survival (% of control)
lambda
kappa
Iota
Mock treated
B
log TCID50
lambda
kappa
iota
Mock treated

Figure 1
Carrageenans promote cell viability of HRV2 infected HeLa cells and inhibit HRV2 replication in vitro.
A. HeLa cells were infected with HRV2 (0,1 TCID50/cell) in the presence of Carrageenans at a concentration of 200 µg/ml. Plates were incubated until cells in the control showed >90% damage. Cell proliferation was determined with an XTT-assay. OD values obtained from mock infected cells (compare x-axis) were set to 100%, and the viability of cells infected in the absence of polymer was set to 0% (y-axis). The bars represent the mean of a quadruplicate experiment, the standard deviation is indicated. B. HeLa cells were infected with HRV2 (0,1 TCID50/cell) in the presence of Carrageenans at a concentration of 200 µg/ml. Viral infectivity in the supernatants was determined by TCID50 assay on HeLa cells (y-axis). Values represent the mean of six parallel titrations, standard deviation is indicated.

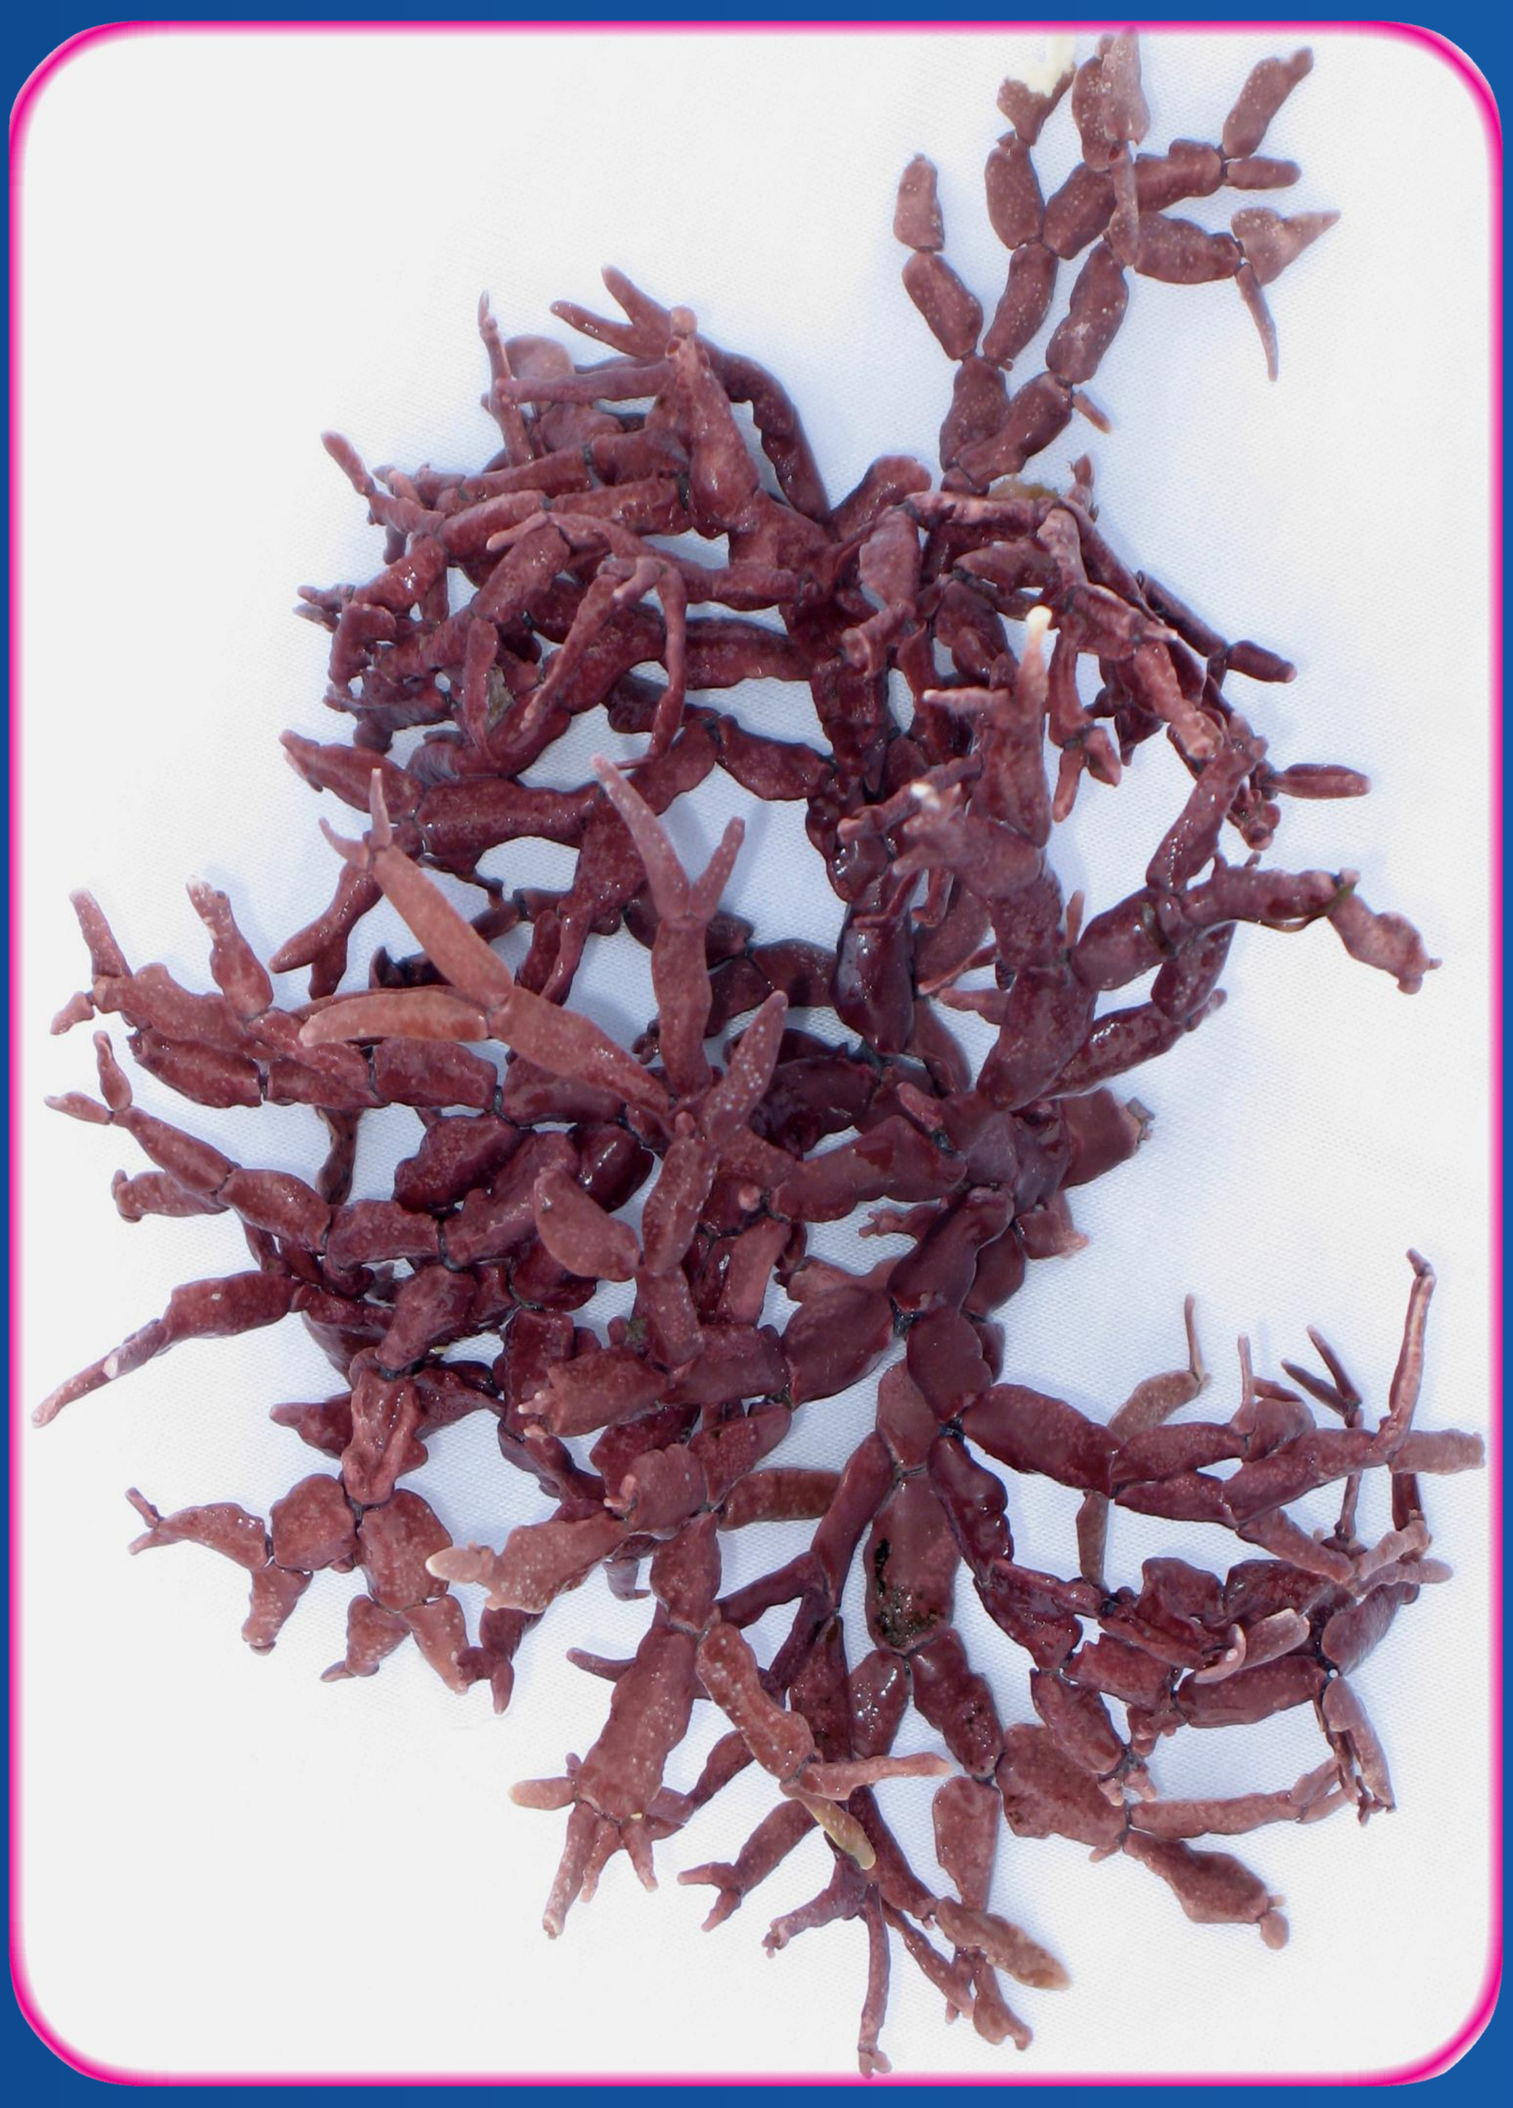

Figure 2
Red seaweed

The anti-viral effect of Iota-Carrageenan is dependent on the amount of input virus

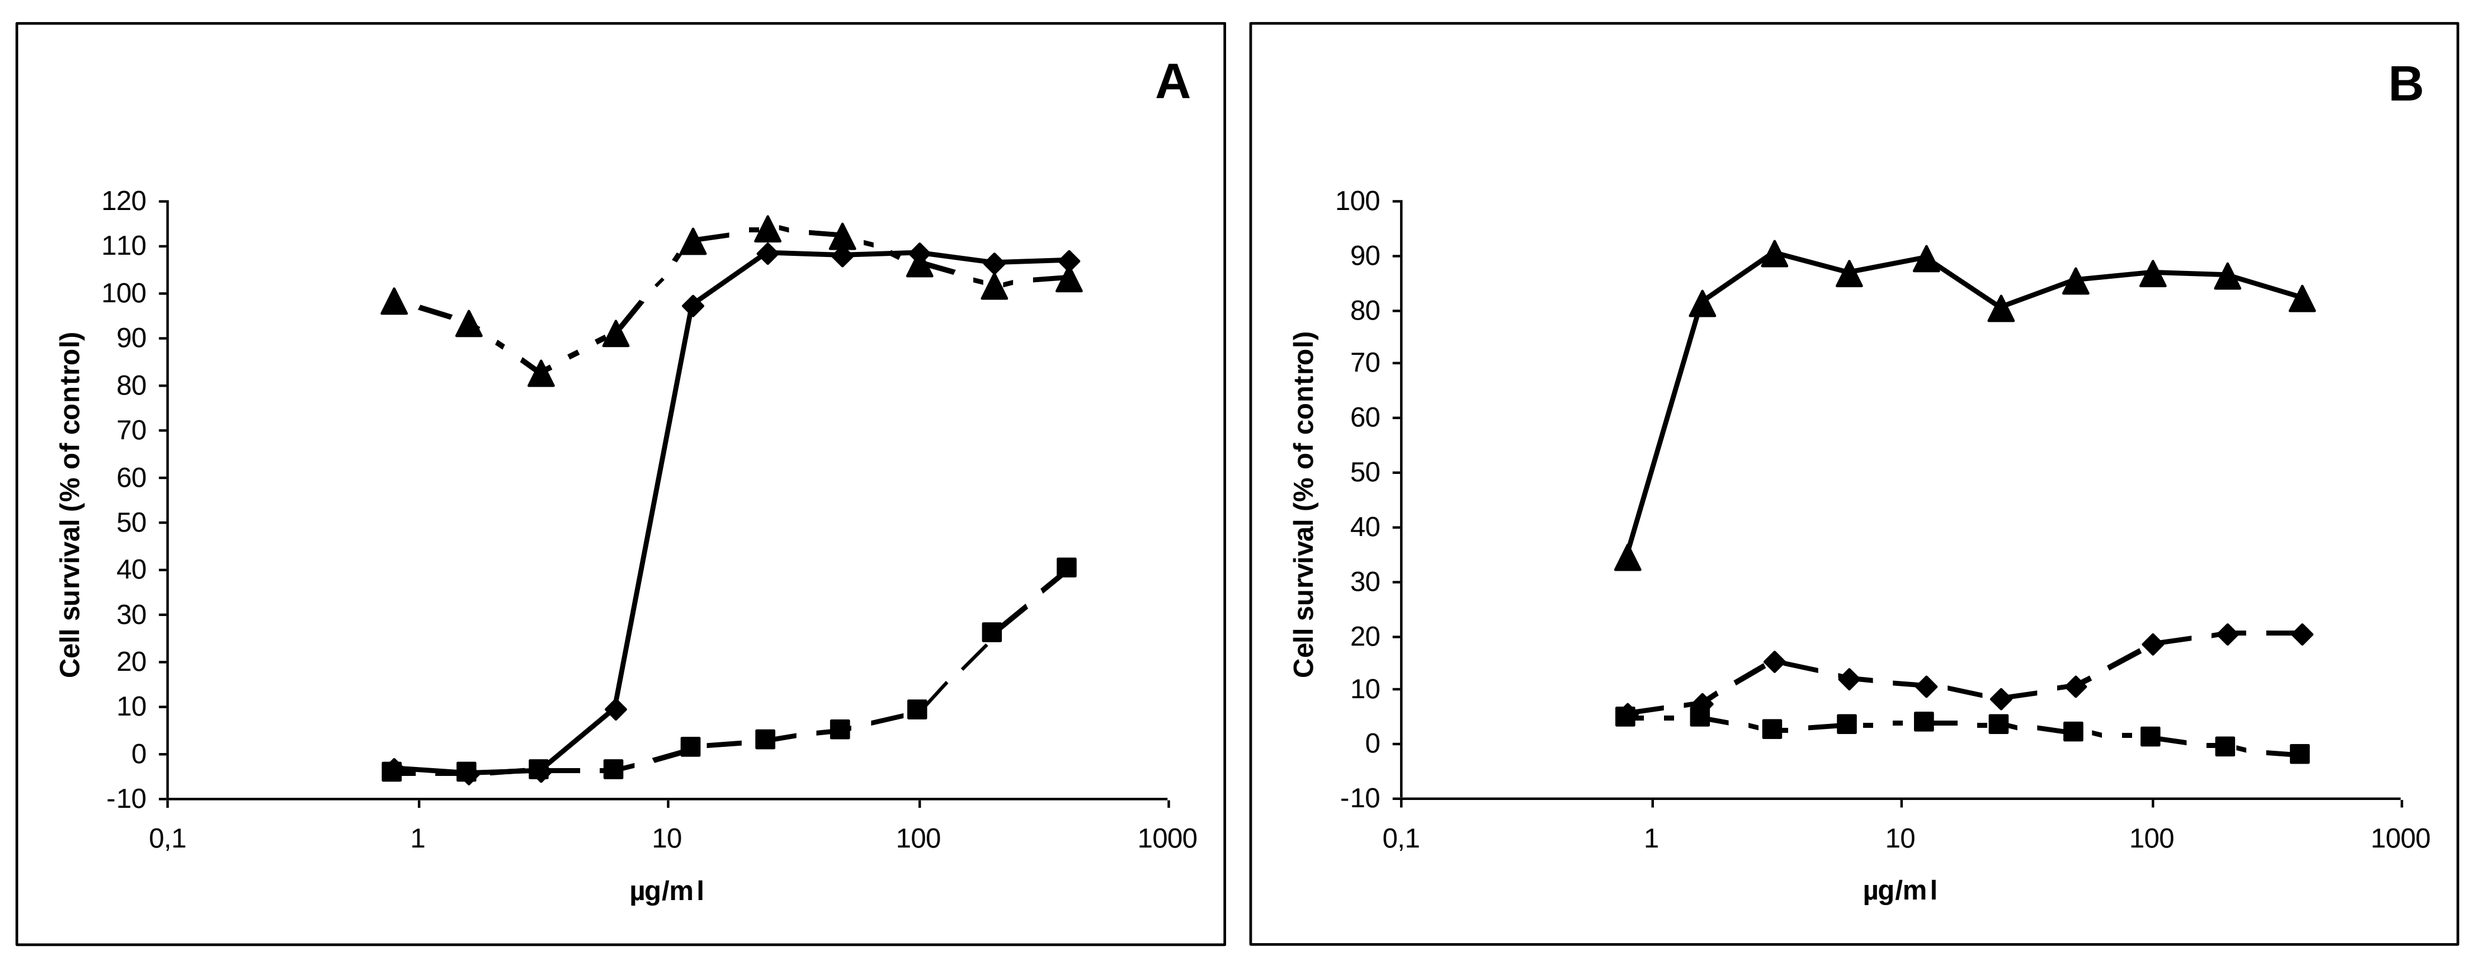
A
Cell survival (% of control)
µg/ml
B
Cell survival (% of control)
µg/ml

Figure 3
Iota-Carrageenan induced inhibition of HRV2 infected cells is dependent on the amount of virus.
A. Preincubation of virus with polymer. HeLa cells were infected with HRV2 in the presence of Iota-Carrageenan. B. Treatment with polymer after infection. HeLa cells were infected with HRV2. 30 minutes after infection medium containing Carrageenan was added. Plates were incubated until cells in the control showed >90% damage. Cell proliferation was determined with an XTT-assay. OD values obtained from mock infected cells (compare x-axis) were set to 100%, and the viability of cells infected in the absence of polymer was set to 0% (y-axis). Black triangles indicate an amount of input virus of 0,01 TCID50/cell, black diamonds indicate 0,1 TCID50/cell and black squares indicate 1 TCID50/cell. A representative experiment is shown

The anti-viral effect of Iota-Carrageenan is dependent on the amount of input virus

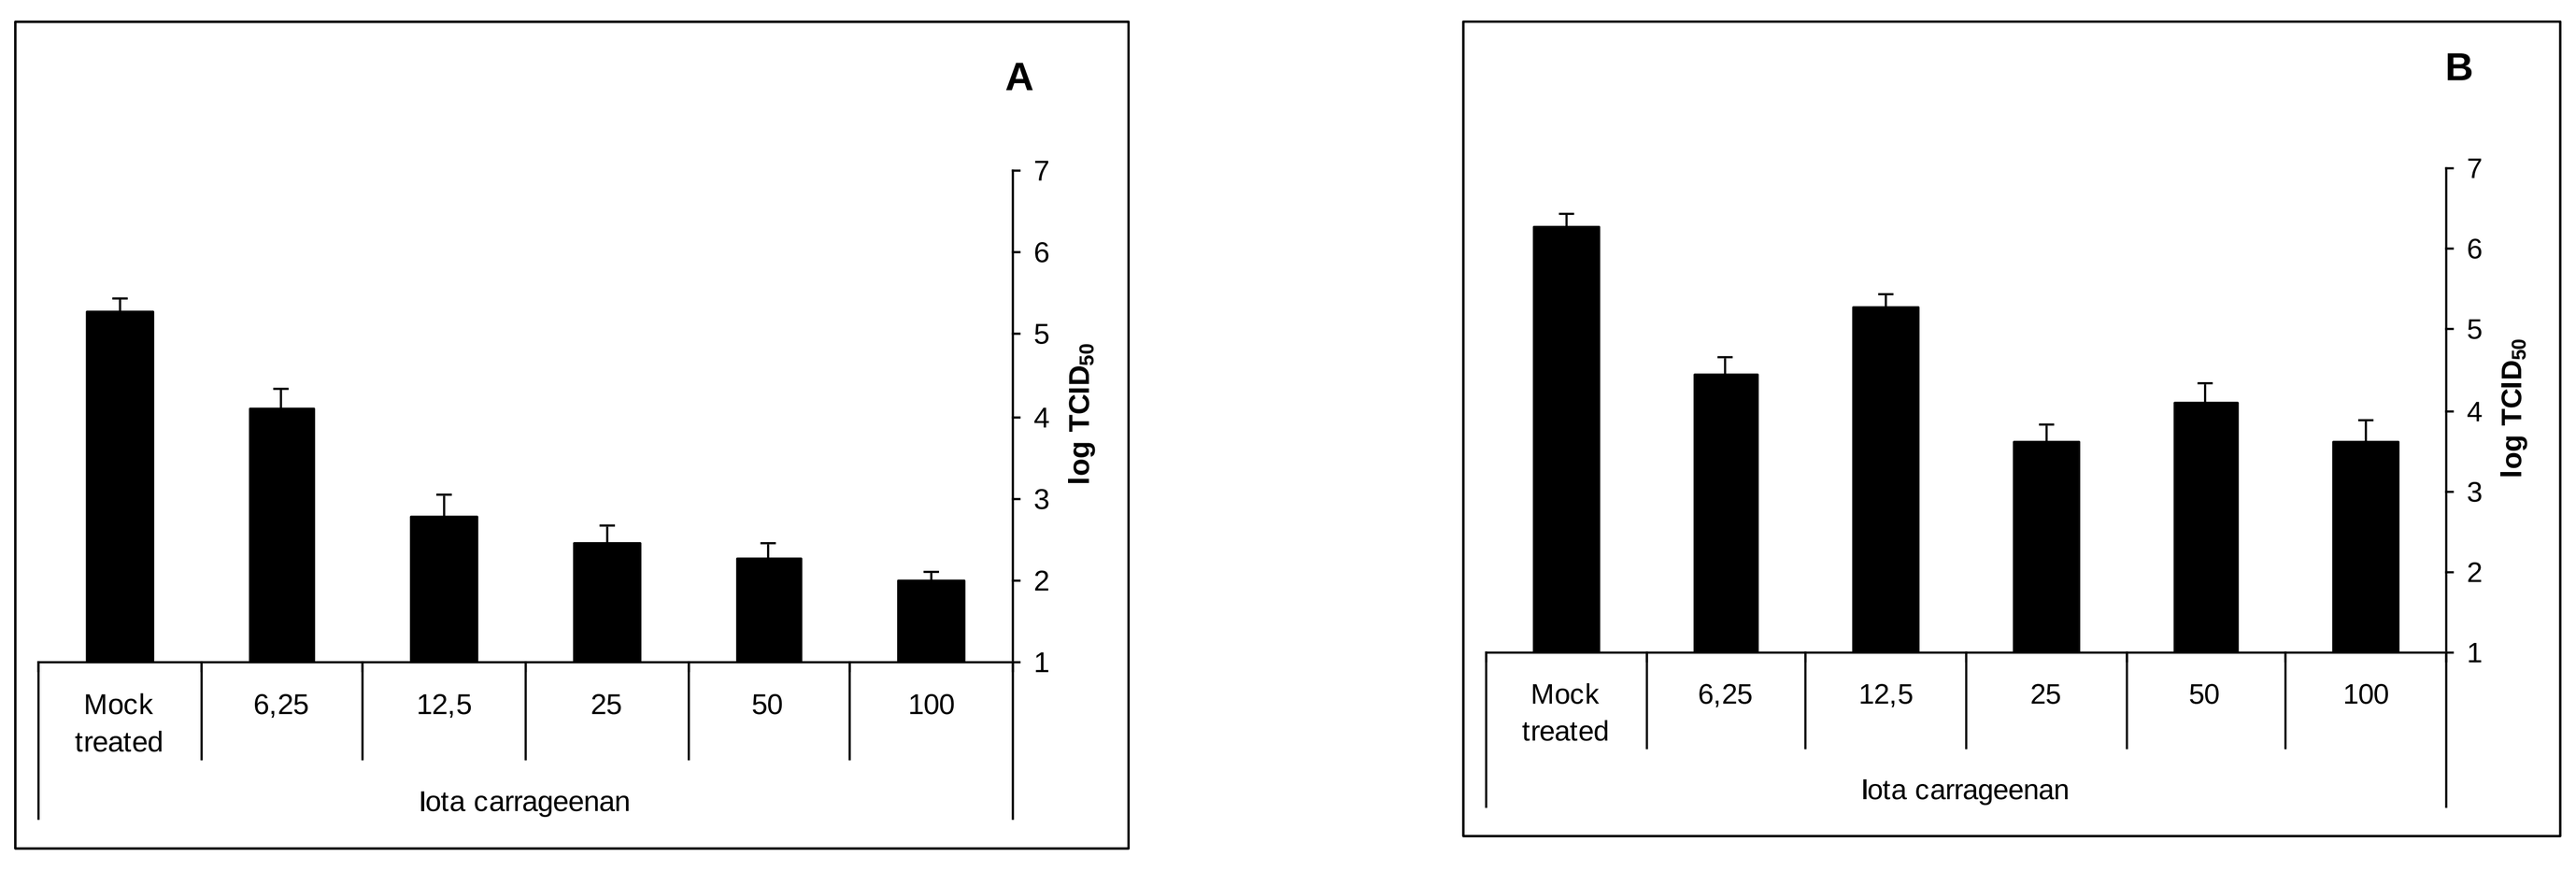
A
log TCID50
Mock treated
6,25
12,5
25
50
100
Iota carrageenan
B
log TCID50
Mock treated
6,25
12,5
25
50
100
Iota carrageenan

Figure 4
Iota-Carrageenan dose-dependently inhibits HRV2 replication in cell culture.
A. Preincubation of virus with polymer. HeLa cells were infected with HRV2 (0,1 TCID50/cell) in the presence of Iota-Carrageenan. 30 minutes after infection the inoculum was removed and medium containing Iota-Carrageenan was added. Untreated cells were used as control).
B. Treatment with polymer after infection. HeLa cells were infected with HRV2 (0,1 TCID50/cell). 30 minutes after infection the inoculum was removed and medium containing Iota-Carrageenan was added. Untreated cells were used as control. Viral titers in the supernatants of infected cells were determined after 48 h by TCID50 assay on HeLa cells. Values are the means from six parallel titrations, standard deviation is indicated.

Iota-Carrageenan does not induce HRV2 escape mutants after 10 passages

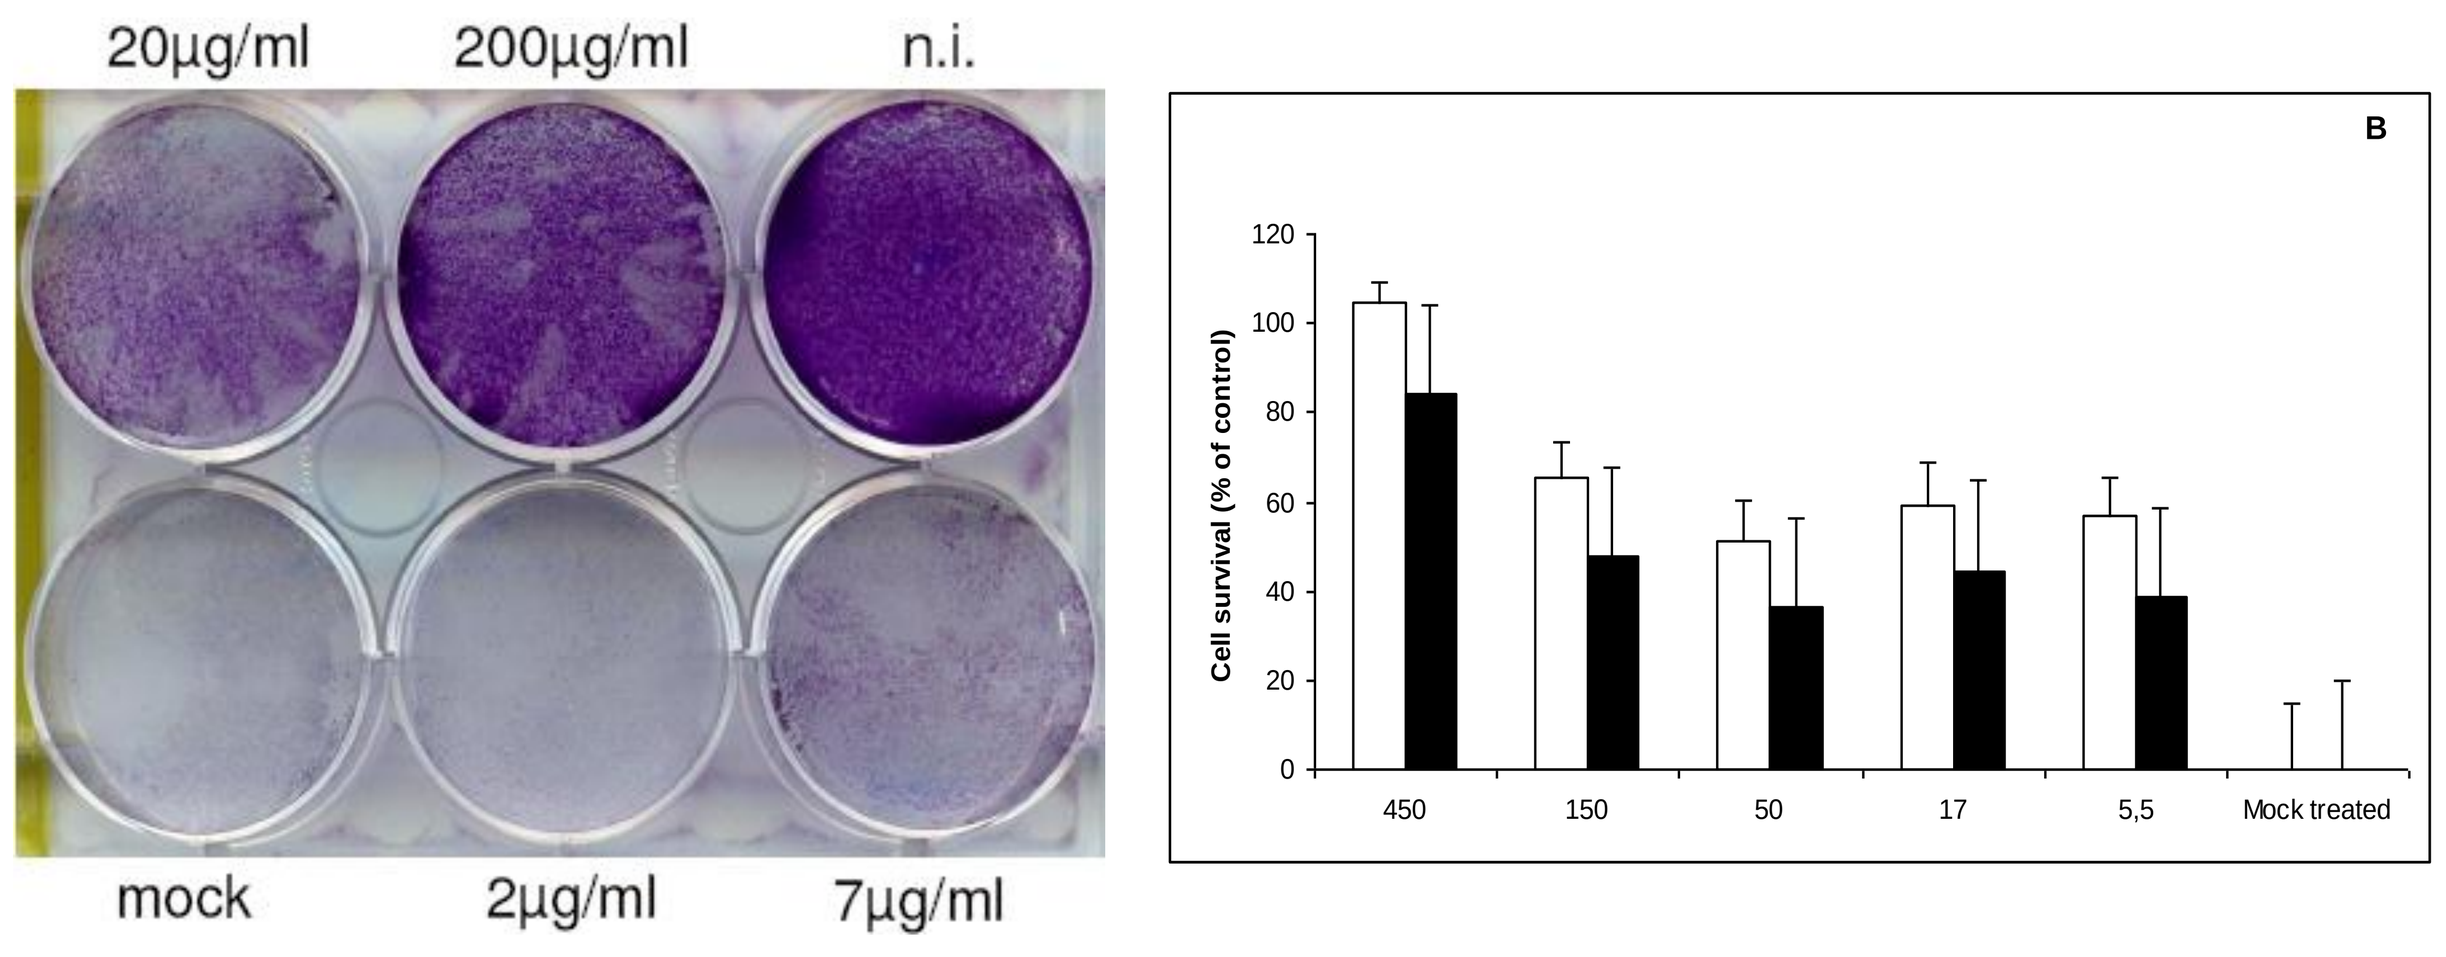
20µg/ml
200µg/ml
n.i.
mock
2µg/ml
7µg/ml
B
Cell survival (% of control)
450
150
50
17
5,5
Mock treated

Figure 5
Search for Iota-Carrageenan resistant variants
A. HeLa cells were infected with HRV2 in the presence of Iota-Carrageenan. Plates were incubated until cells in the control showed >90% damage. B. Supernatants from infected wells were used for the subsequent infection round. After ten repetitive experiments the sensitivity of the resulting virus (white bars) to different concentrations of Iota-Carrageenan (x-axis) was compared with that of the original virus (black bars). Cell proliferation was determined with an XTTassay. Survival of mock infected cells was set to 100%, and that in the absence of polymer was set to 0% (y-axis). The bars represent means of six independent experiments standard deviation is indicated.

Iota-carageenan blocks replication of HRV2 in primary human nasal epithelial cells (HNep)

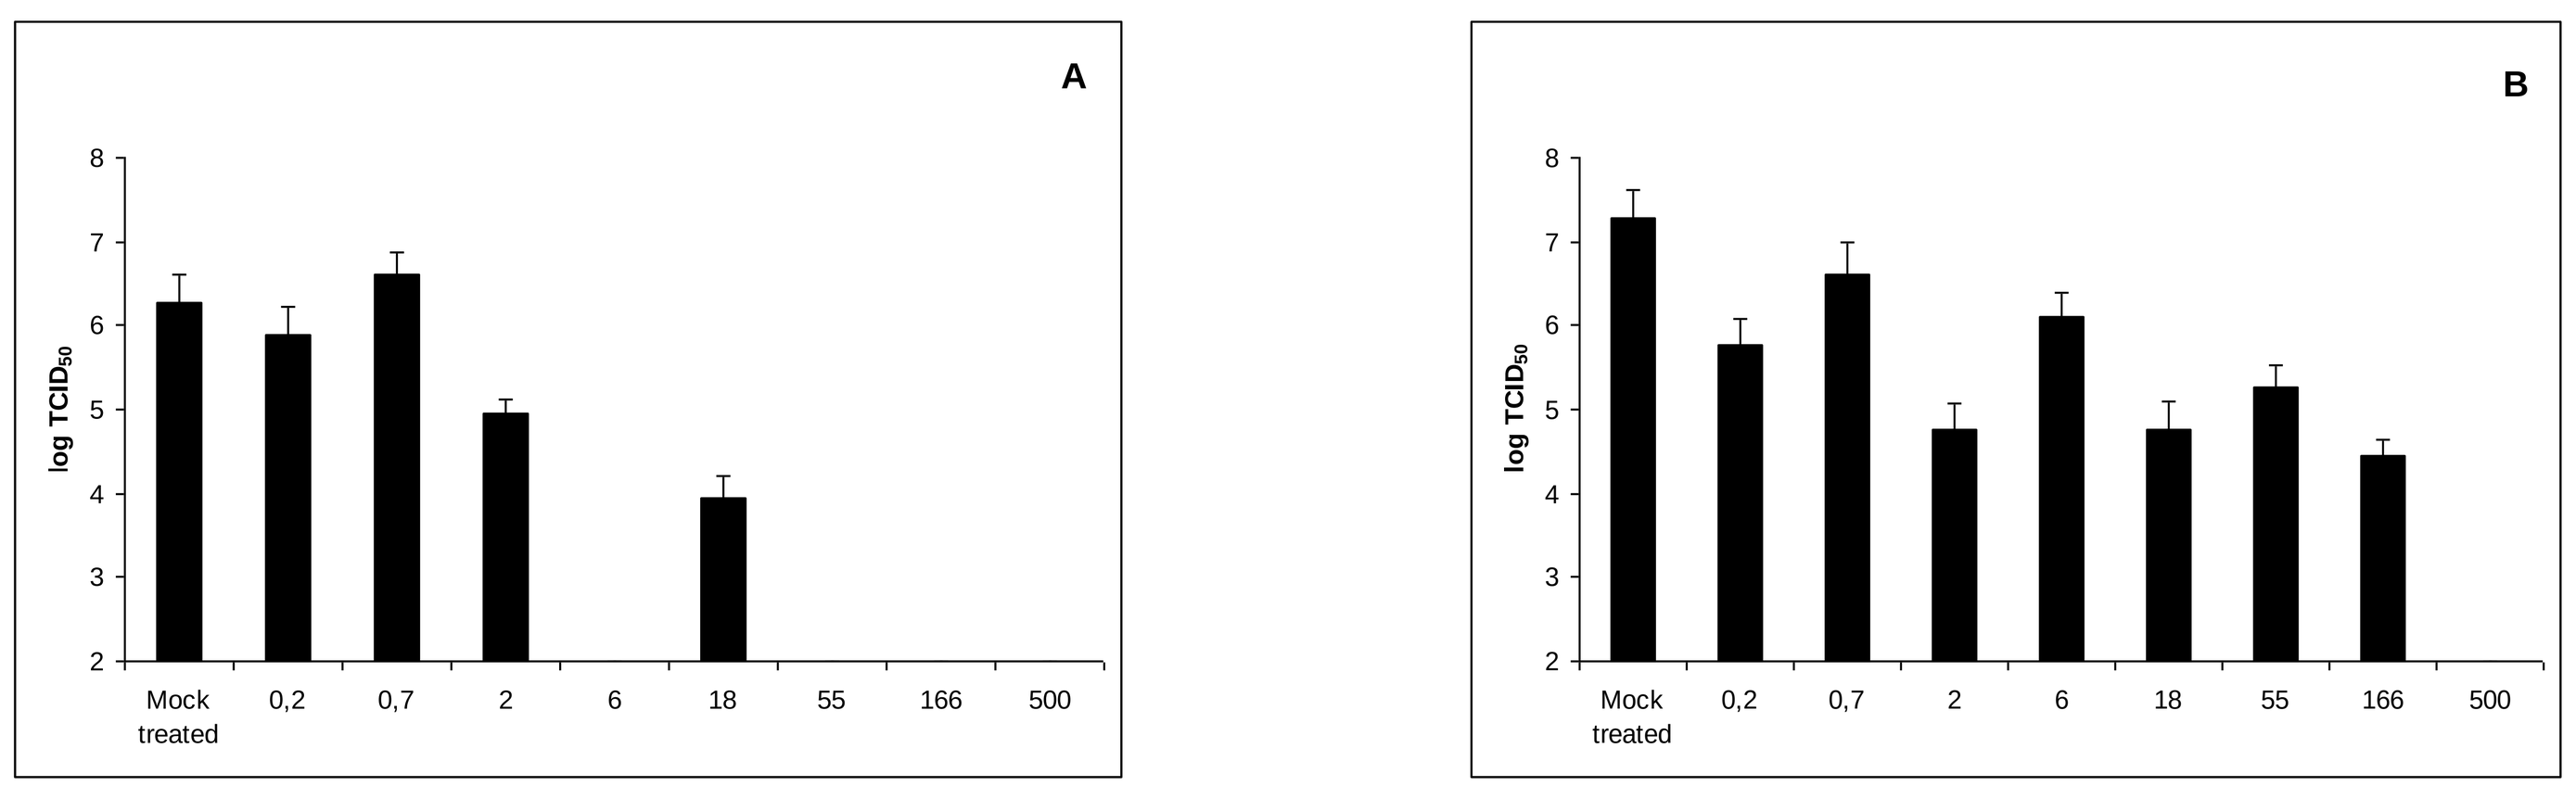
A
log TCID50
Mock treated
0,2
0,7
2
6
18
55
166
500
B
log TCID50
Mock treated
0,2
0,7
2
6
18
55
166
500

Figure 6
Effect of Iota-carageenan on HRV2 infected human nasal epithelial cells.
A. Preincubation of virus with polymer. HNep cells were infected with HRV2 (0,1 TCID50/cell) in the presence of Iota-Carrageenan. 30 minutes after infection the inoculum was removed and medium containing Iota-Carrageenan was added. B. Treatment with polymer after infection. HNep cells were infected with HRV2 (0,1 TCID50/cell). 30 minutes after infection the inoculum was removed and medium containing Iota-Carrageenan was added. Viral titers in the supernatants of infected cells were determined after 48 h by TCID50 assay on HeLa cells (y-axis). Bars represent means of four parallel experiments,standard deviation is indicated.

Iota-carrageenan inhibits replication of HRV serotypes 1A, 8, 14, 16 and 83 on primary human epithelial cells

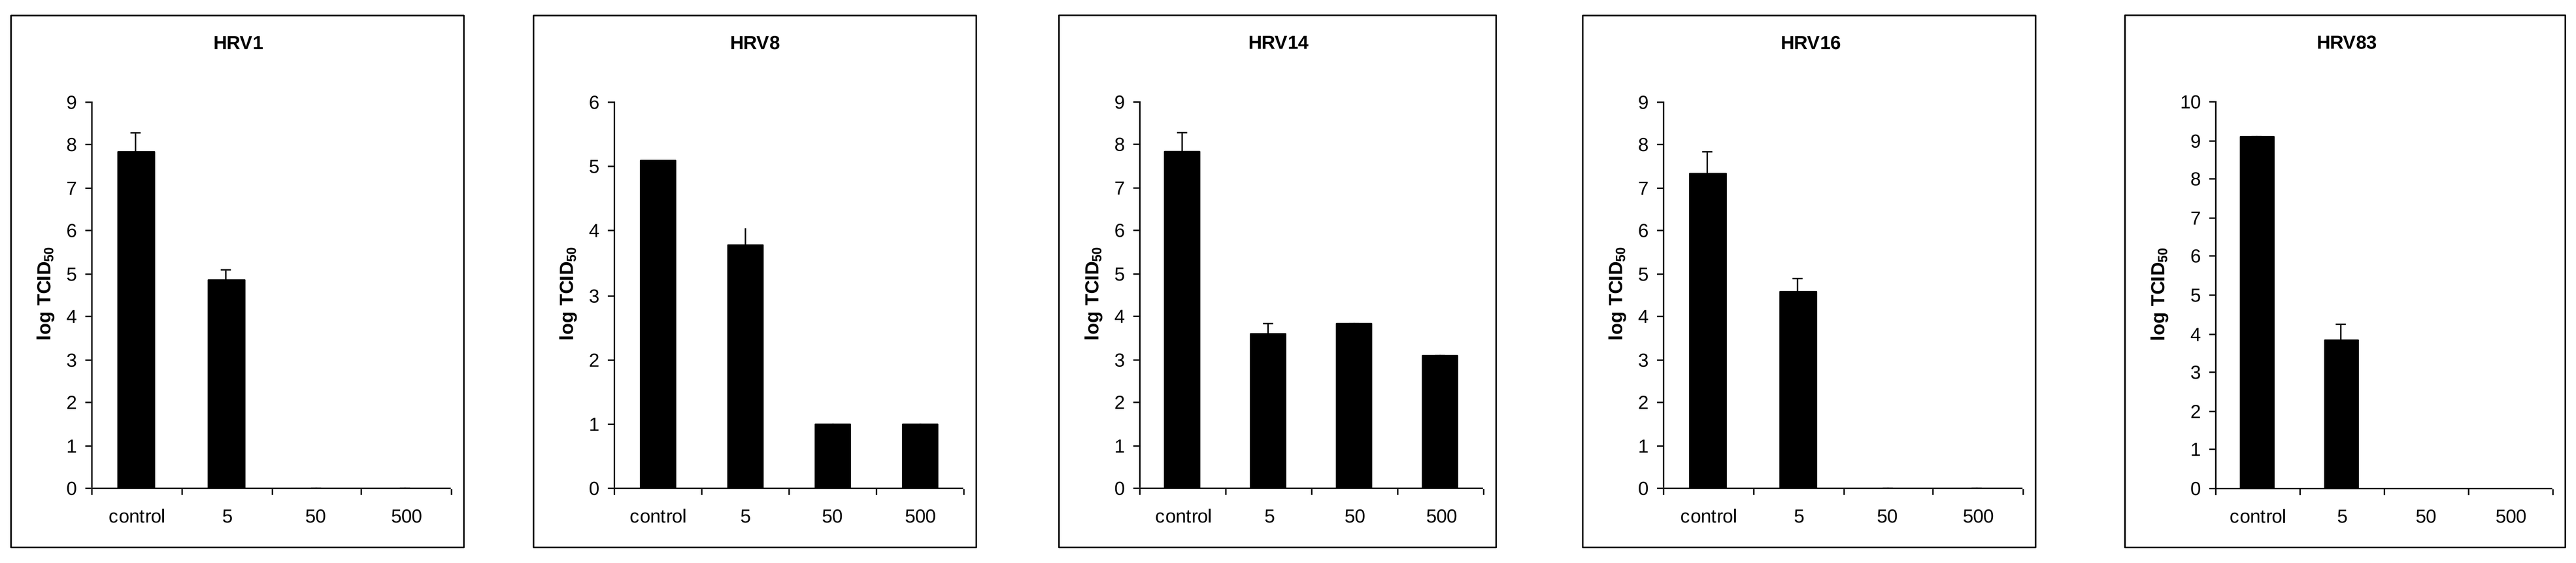
HRV1
HRV8
HRV14
HRV16
HRV83
log TCID50
control
5
50
500

Figure 7
Effect of Iota carageenan on the replication of HRV strains 1A, 8, 14, 16 and 83 on human nasal epithelial cells.
HNep cells were infected with different HRV strains (indicated at the top of each panel; 0,1 TCID50/cell) in the presence of Iota-Carrageenan at the concentrations indicated at the x-axis. 30 minutes after infection the inoculum was removed and medium containing Iota-Carrageenan was added. Viral titers in the supernatants of infected cells were determined after 48 h by TCID50 assay on HeLa cells (y-axis). Bars represent means from four parallel experiments, standard deviations are indicated.

Reference: Grassauer A, Weinmuellner R, Meier C, Prieschl-Grassauer E, Unger H: **Iota-Carrageenan is a potent inhibitor of rhinovirus infection.** *Virology Journal* 2008, **5:**107