Contact: martina.kurz@marinomed.com

Combinational antiviral therapy with Carragelose® and Zanamivir is synergistically active against influenza A virus infection

M. Morokutti-Kurz, S. Nakowitsch, M. König-Schuster, C. Graf, C. Koller, N. Kirchoff, A. Grassauer, E. Prieschl-Grassauer

Acute viral respiratory tract infections (ARTI) are a global burden to public health. ARTI are caused by single or mixed infection with various respiratory viruses, including rhino- (hRV), corona-, respiratory syncytial, and influenza viruses (IV). Therefore, a broad antiviral therapy is needed. A nasal spray containing Carragelose® has shown antiviral effectiveness against a broad variety of respiratory viruses in several clinical trials. Zanamivir, a neuraminidase inhibitor, is effective against IV infections also when applied intranasally. A potential synergistic antiviral activity of a therapy consisting of Carragelose® and Zanamivir was evaluated.

In vitro the antiviral effectiveness of Carragelose® against hRV is maintained in the combination formulation. Compared to Zanamivir alone, the combination product is 50 times (H7N7), two (H1N1/09pdm), three (H5N1) or even five (H3N2) orders of magnitudes more potent in inhibiting IV infection. This synergistic effect was confirmed in vivo. Lethal mouse-models were established for IV A H1N1/09pdm and H7N7 infections. Mice were treated intranasally two times a day for five days with Carragelose®, Zanamivir or the combination product. For both IV subtypes the survival rate of mice was significantly higher for the combination product compared to Zanamivir alone. The therapy was effective even when the treatment started as late as 48 (H7N7) or 72 (H1N1/09pdm) hours after infection.

We conclude that intranasal administration of the combination product is a promising option for the prophylaxis and therapy of ARTI.

Introduction

Monotherapy - Carrageenan

Carrageenan is a **sulfated polymer** derived from red seawead. The intranasal application of carrageenan creates a **protective physical barrier** in the nasal cavity and works as inhibitor against virus entry. Its antiviral effectiveness was confirmed towards various respiratory viruses in several clinical trials.

Monotherapy - Zanamivir

Two **neuraminidase (NA) inhibitors**, Oseltamivir and Zanamivir, are available on the market for treatment and prophylaxis of IV infections. However, increasing use of Zanamivir monotherapy increases the risk of developing resistance. Zanamivir is directly applied as active drug and can be administered intranasally.

Combinational therapy - Carragelose® and Zanamivir

IV infections are often accompanied by infections with other viral pathogens. As a result, the therapy with an anti-influenza monotherapy often fails to sufficiently resolve symptoms. The combination of Zanamivir, which reliably treats IV infections, and Carragelose®, active against a broad range of respiratory viruses, can overcome this problem.

Zanamivir and Carragelose® have fundamentally different mechanisms of action and different targets. As a result, the risk of developing resistance is minimized while at the same time full protection against various IV strains and respiratory viruses is privided.

Results – *in vitro*

The Antiviral effectiveness of Carragelose® is not affected by the presence of Zanamivir and vice versa

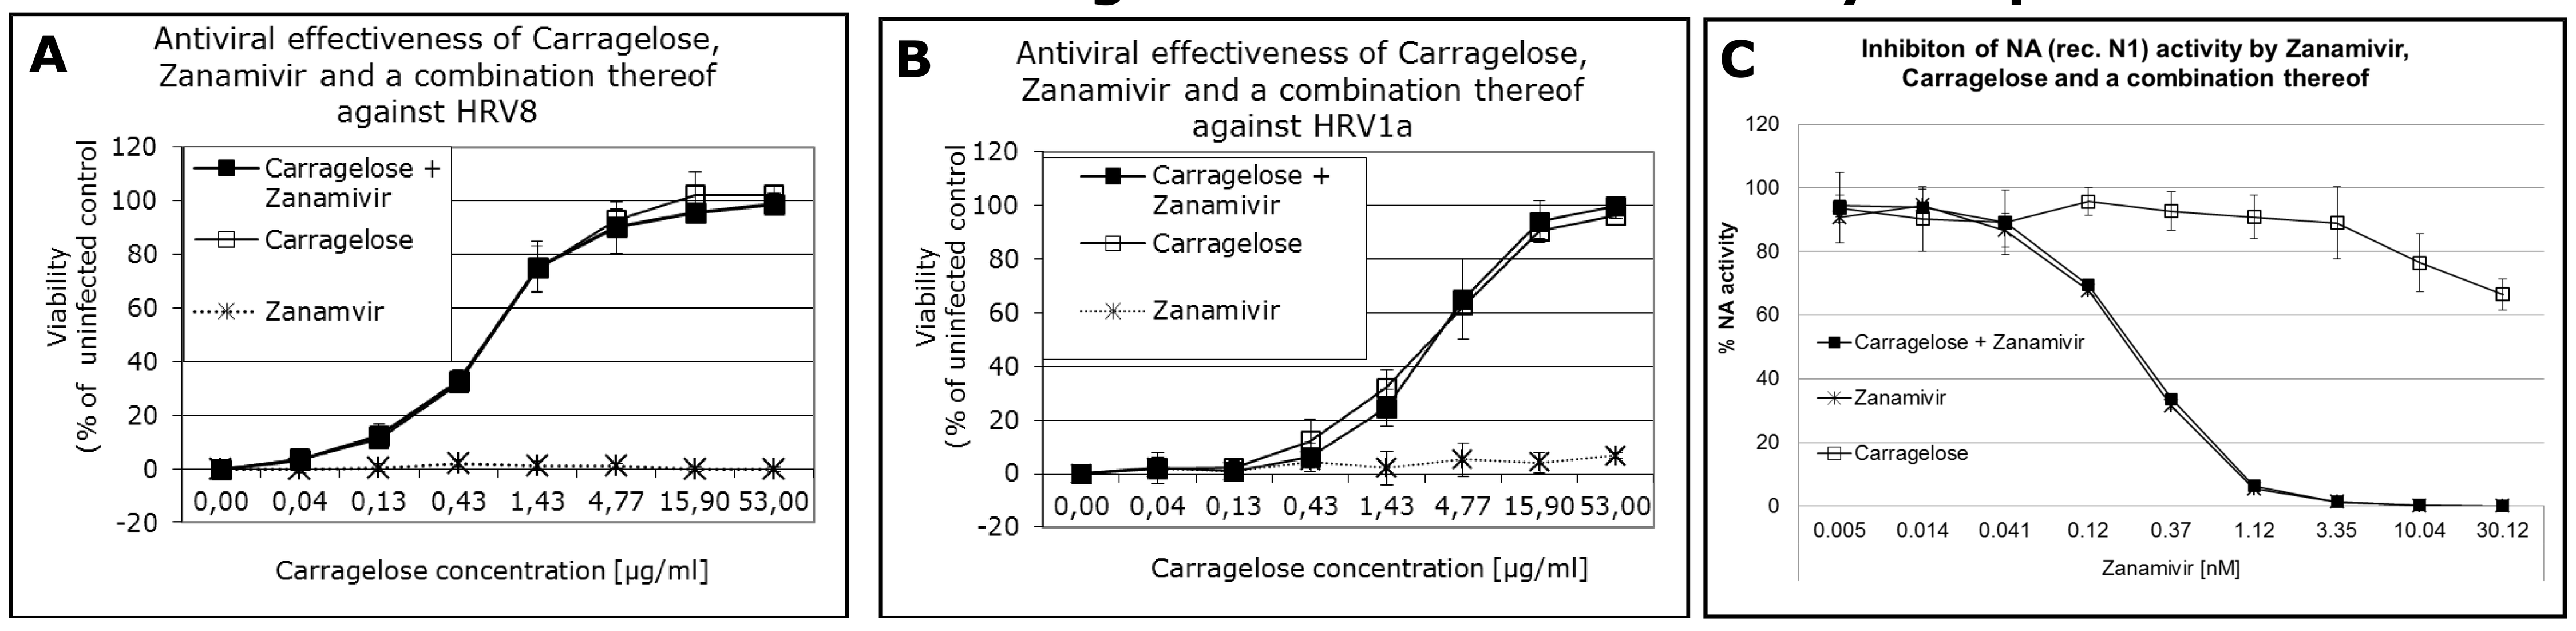
A
Antiviral effectiveness of Carragelose, Zanamivir and a combination thereof against HRV8
Viability (% of uninfected control)
Carragelose + Zanamivir
Carragelose
Zanamvir
Carragelose concentration [µg/ml]
B
Antiviral effectiveness of Carragelose, Zanamivir and a combination thereof against HRV1a
Viability (% of uninfected control)
Carragelose + Zanamivir
Carragelose
Zanamivir
Carragelose concentration [µg/ml]
C
Inhibiton of NA (rec. N1) activity by Zanamivir, Carragelose and a combination thereof
% NA activity
Carragelose + Zanamivir
Zanamivir
Carragelose
Zanamivir [nM]

The anti-viral efficacy of Carragelose®, Zanamivir and a combination thereof against hRV was tested using a CPE reduction assay on HeLa cells. Cell survival was measured using a resazurin based metabolic assay. The graphs show the mean cell viability as percentage of the uninfected control and the standard deviations of sixtuplicates (A + B). The inhibition of the NA activity of a recombinant N1 NA by Zanamivir in the presence and absence of Carragelose® was tested using a chemiluminescent substrate. The graphs show the mean NA activity of triplicates as percentage of the NA activity of an untreated control (C).

Compared to Zanamivir alone, the combinational therapy is much more potent in inhibiting replication of various IV A strains

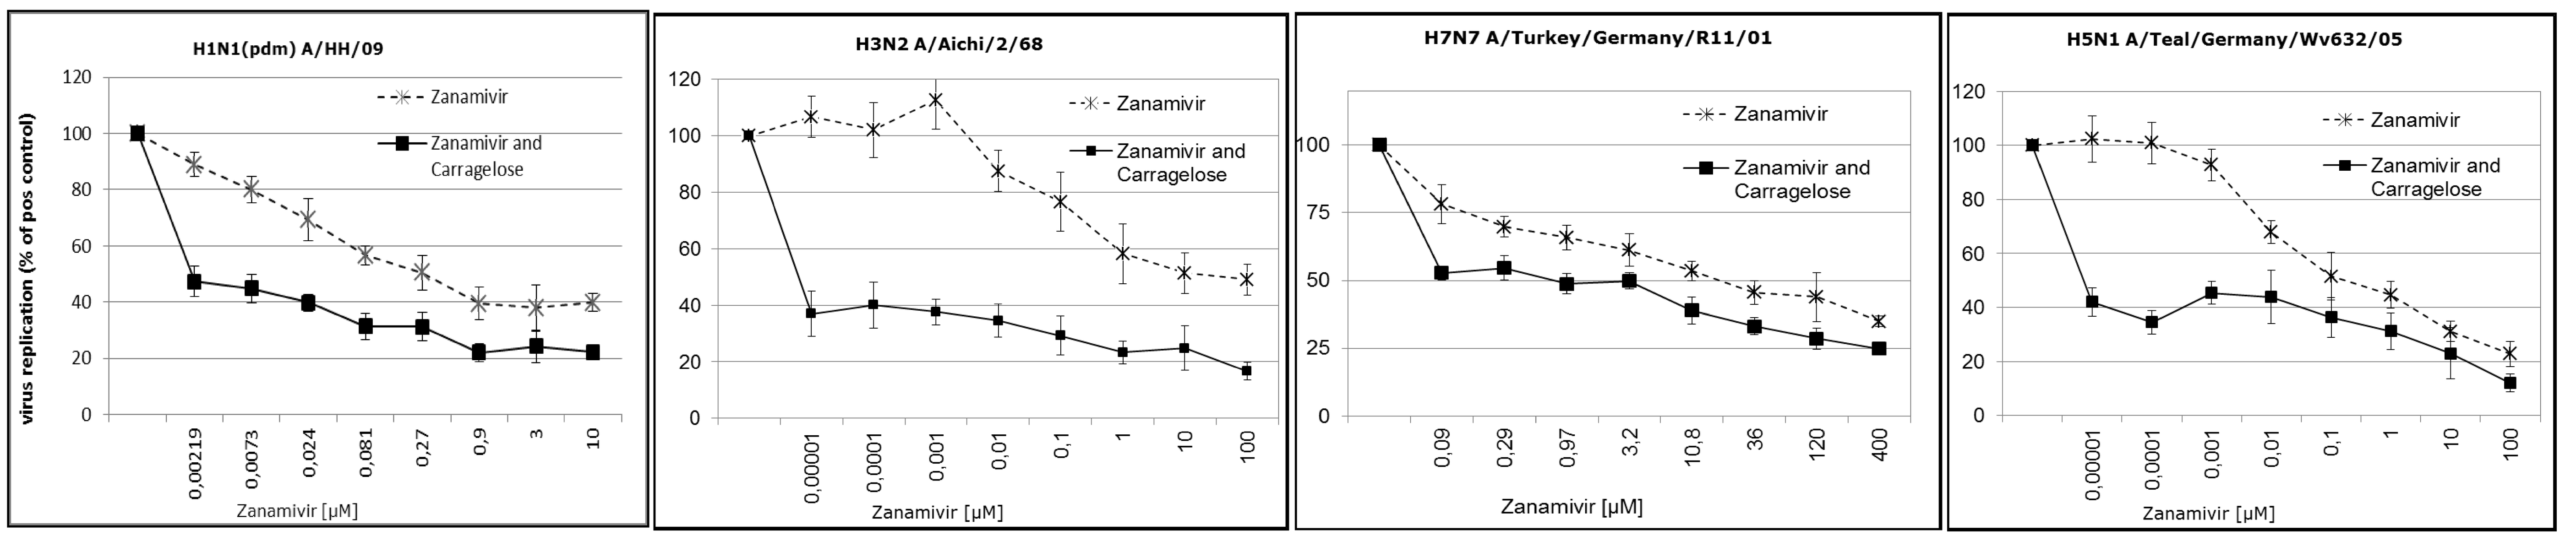
H1N1(pdm) A/HH/09
H3N2 A/Aichi/2/68
H7N7 A/Turkey/Germany/R11/01
H5N1 A/Teal/Germany/Wv632/05
virus replication (% of pos control)
Zanamivir
Zanamivir and Carragelose
Zanamivir [µM]

The **anti-viral** efficacy of Zanamivir and the combination formulation against various **influenza virus** strains was evaluated using a novel plaque reduction assay with a semi-liquid overlay in a 96 well format on MDCK cells. Cells were infected in the presence of the respective antiviral. After incubation, cells were fixed and viral replication was quantified using immuno-staining and a microplate reader.

Results – *in vivo*

Intranasal therapy with Carragelose® and Zanamivir protects lethally influenza infected mice even after delayed treatment start

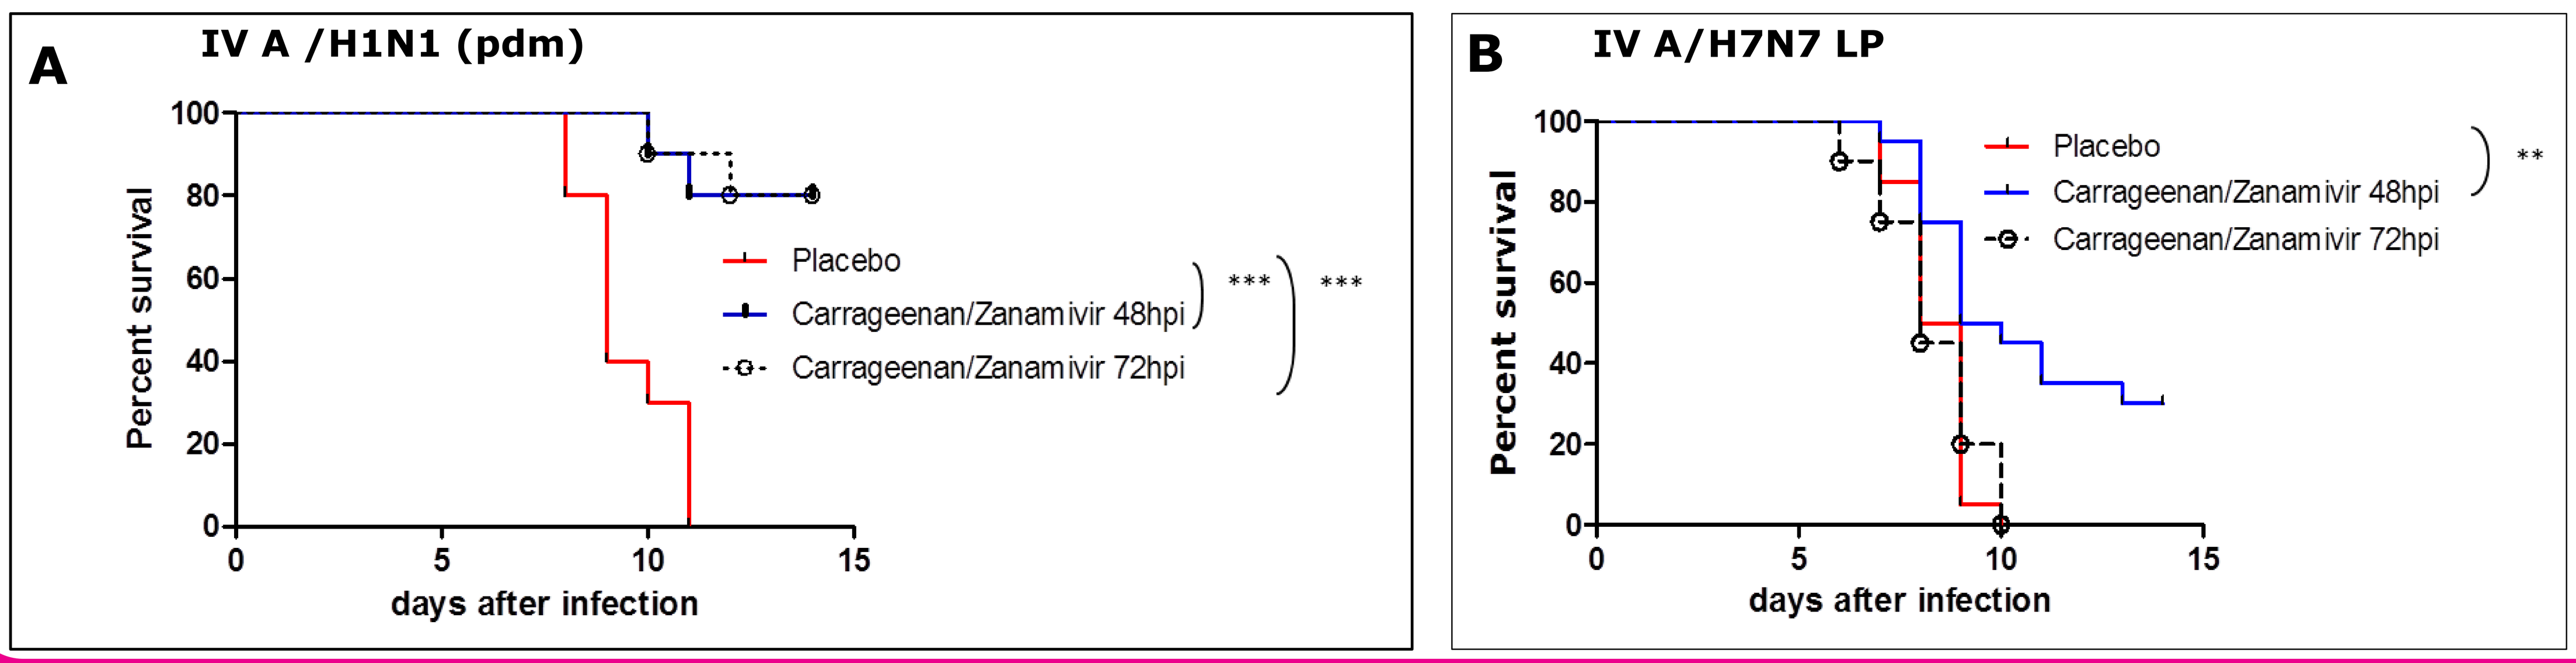
A
IV A /H1N1 (pdm)
Percent survival
Placebo
Carrageenan/Zanamivir 48hpi
Carrageenan/Zanamivir 72hpi

days after infection
B
IV A/H7N7 LP
Percent survival
Placebo
Carrageenan/Zanamivir 48hpi
Carrageenan/Zanamivir 72hpi
**
days after infection

Ten mice per group were intranasally infected with influenza A/HH/01/2009 (A) or H7N7 A/Teal/Germany/Wv632/05 (B) at day 0 and treated intranasally twice a day with placebo or a combinational treatment consisting of Carragelose® and Zanamivir (1mg/kg bodyweight). Treatment started 48 and 72 hours post infection (hpi) and continued for 5 days.

P values were calculated by log rank test
** $p<0.01$
*** $p<0.001$)

Summary

IV infections are often accompanied by infections with other viral pathogens; therefore, anti-influenza monotherapy fails to fight these mixed infections. Furthermore, Zanamivir monotherapies enhance the risk of developing escape mutants. A combinational therapy consisting of Carragelose® and Zanamivir is suitable to fight both problems. We showed that with the combinational therapy a broader range of respiratory viruses is targeted and that the efficiency of both monotherapies is not affected by the respective other compound. Furhermore, we demonstrated that the combination formulation reduces viral replication in cells more efficently than Zanamivir treatment alone. Impressively, intranasal therapy with the combinational therapy protects influenza infected mice from death even when treatment starts as late as 48 and 72 hpi. We conclude that the combinational therapy deserves further investigation in clinical trials.

Reference: 1. Grassauer, A., Weinmüllner, R., Meier, C., Pretsch, A., Prieschl-Grassauer, E., Unger, H. 2008, "Iota-Carrageenan is a potent inhibitor of rhinovirus infection", Virol.J., Vol. 5, No. 107
2. Leibbrandt, A., Meier, C., König-Schuster, M., Weinmüllner, R., Kalthoff, D., Pflugfelder, B., Graf, P., Frank-Gehrke, B., Beer, M., Fazekas, T., Unger, H., Prieschl-Grassauer, E., Grassauer, A. 2010, "Iota-carrageenan is a potent inhibitor of influenza A virus infection", PLoS.One